Dissecting Bcl-2 Sensitivity in AML: A QSP Model for Venetoclax-Azacitidine Therapy

Matthias Hanke[1], Satoshi Nakayama[2], Tomohisa Nakada[2], **Claire Couty**[1], Simon Arsène[1]

[1] Nova In Silico, Lyon, France, [2] Mitsubishi Tanabe Pharma Corporation, Yokohama, Japan

BACKGROUND

Acute Myeloid Leukemia (AML) is an aggressive blood cancer affecting the myeloid lineage of hematopoiesis. Despite significant strides in treatment, it remains a challenge for older patients or those unfit for intensive chemotherapy regimens. A standard of care therapy for such patients involves combining Venetoclax (VEN), inhibiting the anti-apoptotic Bcl-2 protein, with Azacitidine (AZA), a hypomethylating agent. This combination disrupts the growth and survival of cancer cells, offering a well-established and effective therapeutic approach for AML patients. However, despite the success of VEN-AZA therapy, individual responses vary significantly, and some patients eventually relapse. Understanding the reasons behind this variability is crucial for improving treatment outcomes.

New mechanistic model sheds light on how Bcl-2 could explain **response stratification in AML VEN-AZA** therapy

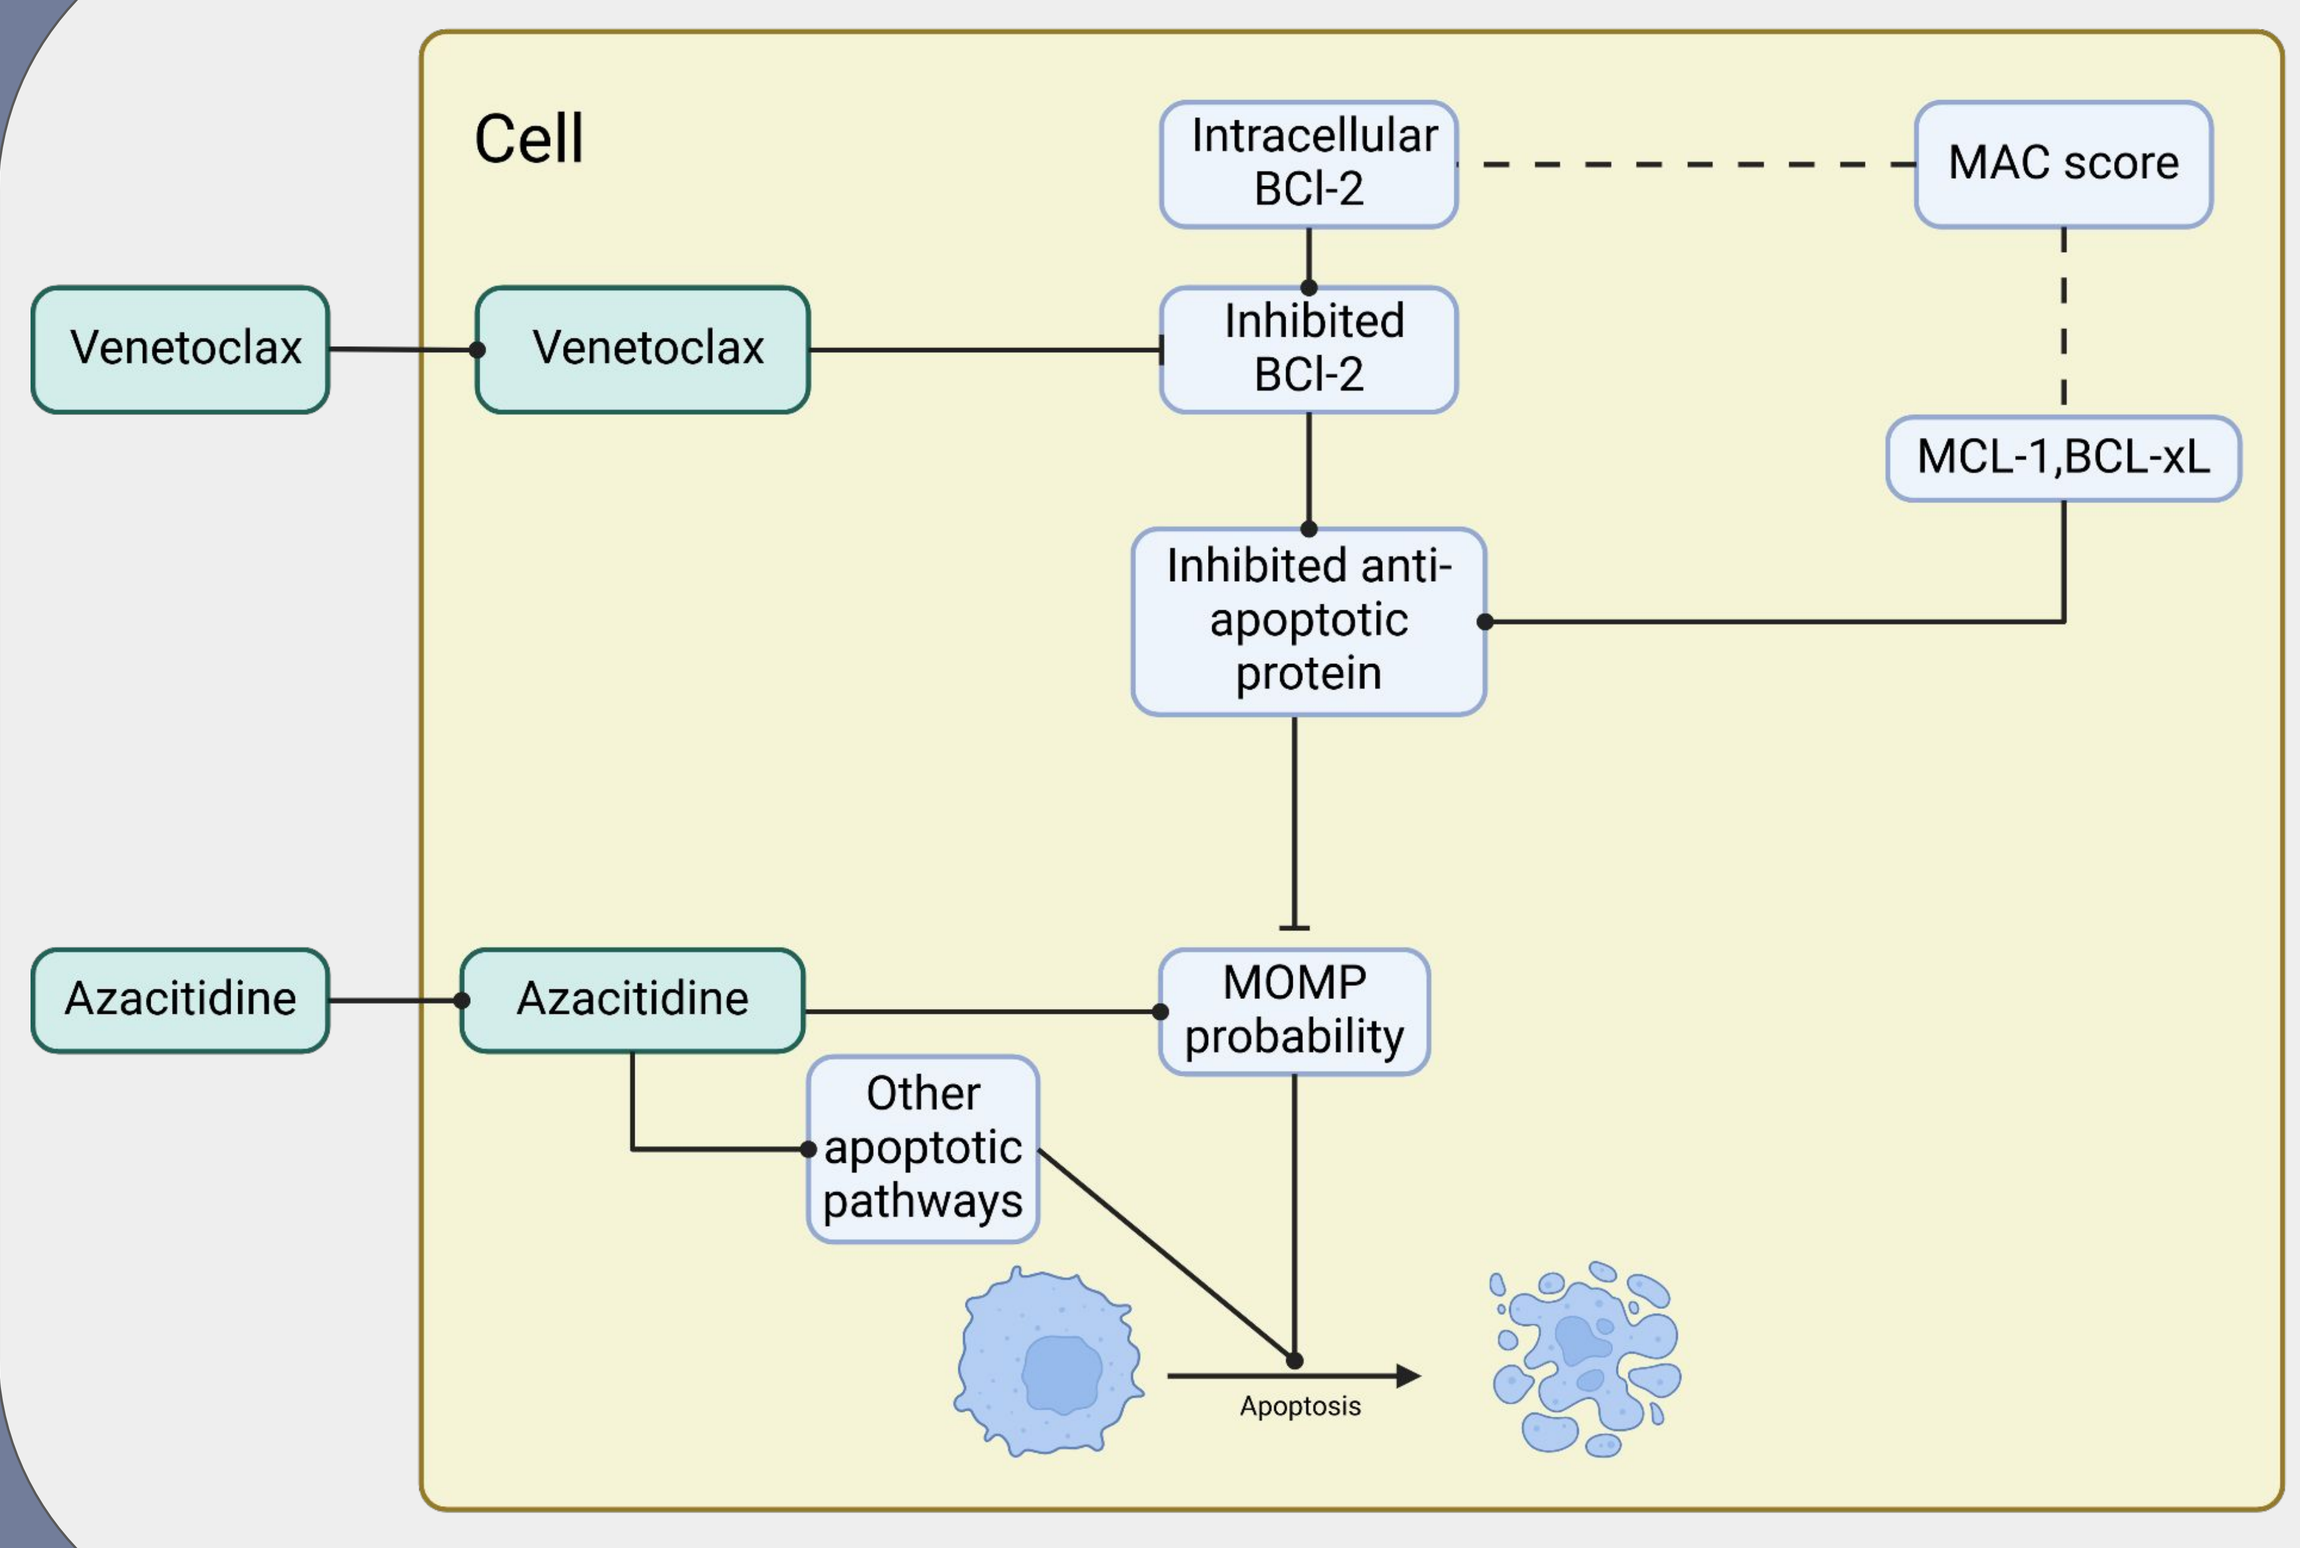
Cell
Venetoclax
Venetoclax
Intracellular BCl-2
Inhibited BCl-2
MAC score
MCL-1,BCL-xL
Inhibited anti-apoptotic protein
Azacitidine
Azacitidine
MOMP probability
Other apoptotic pathways
Apoptosis

Figure 1: Apoptosis submodel's graph including main entities and treatment effects.

MOMP: Mitochondrial outer membrane permeabilization leading to apoptosis.

MAC score: Mediators of apoptosis combinatorial score, defined as the ratio of Bcl-2 expression over other anti-apoptotic protein expression (BCL-xL and MCL)[1]

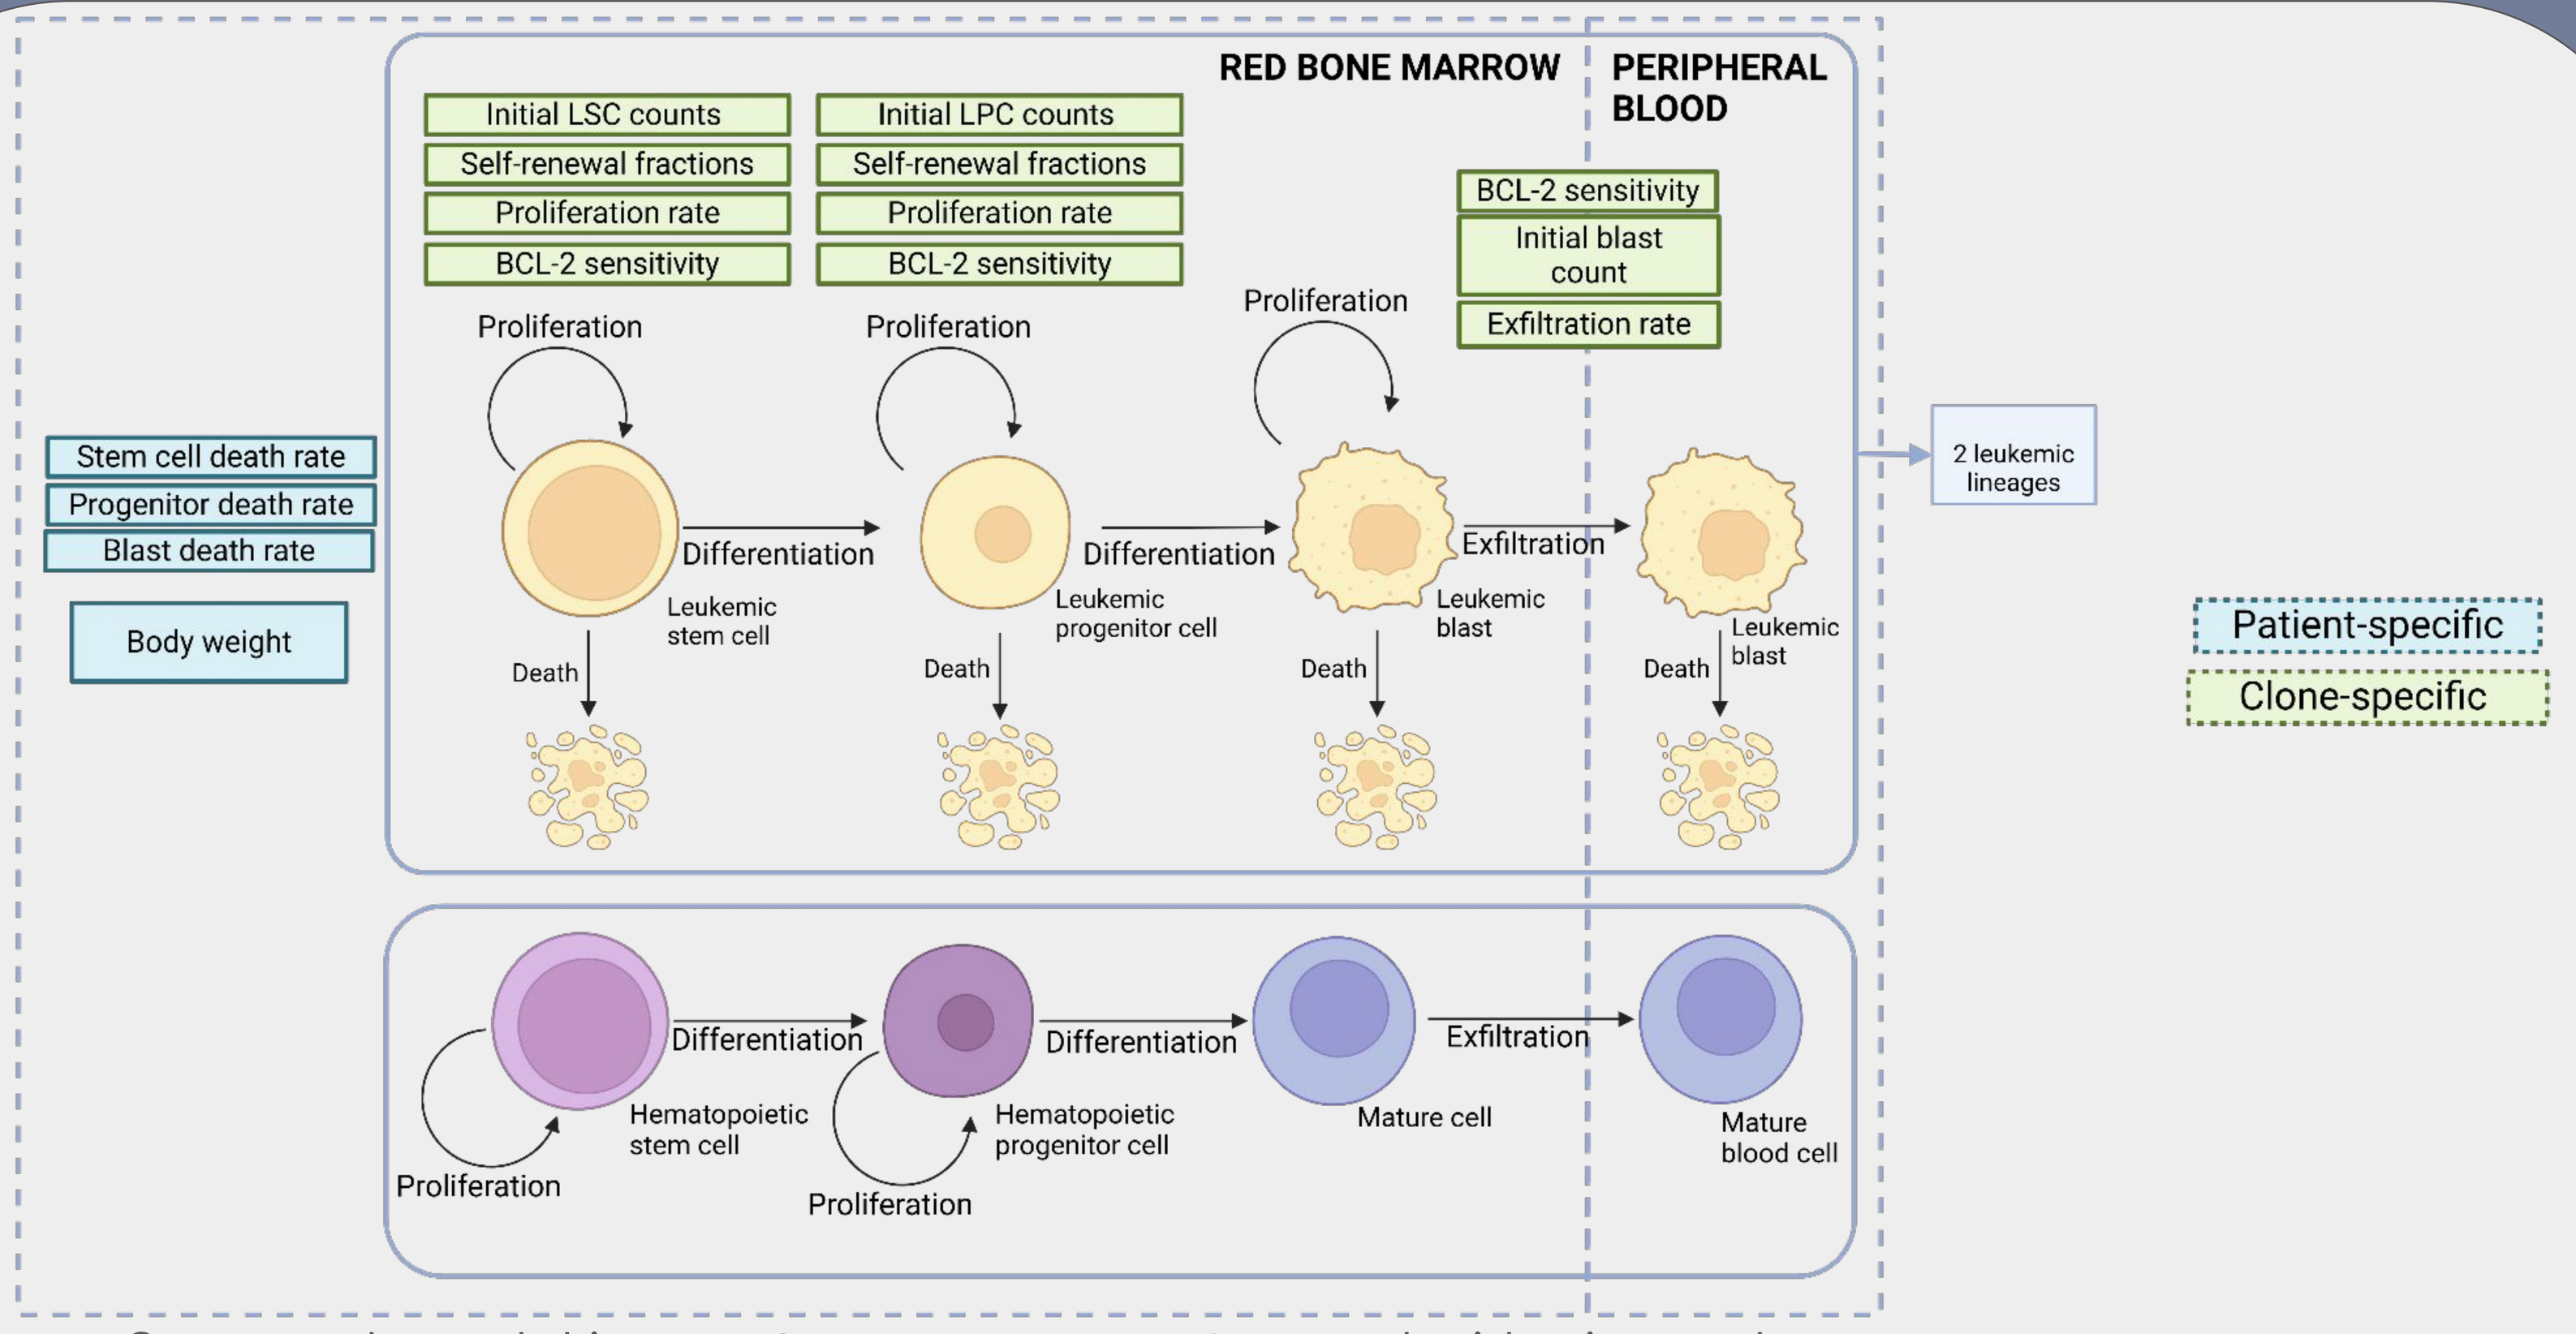
RED BONE MARROW
PERIPHERAL BLOOD
Initial LSC counts
Self-renewal fractions
Proliferation rate
BCL-2 sensitivity
Initial LPC counts
Self-renewal fractions
Proliferation rate
BCL-2 sensitivity
BCL-2 sensitivity
Initial blast count
Exfiltration rate
Stem cell death rate
Progenitor death rate
Blast death rate
Body weight
Proliferation
Differentiation
Leukemic stem cell
Leukemic progenitor cell
Leukemic blast
Exfiltration
Death
2 leukemic lineages
Patient-specific
Clone-specific
Hematopoietic stem cell
Hematopoietic progenitor cell
Mature cell
Mature blood cell

Figure 2: AML submodel inputs & parameters - Created with BioRender.com

A multistep calibration process

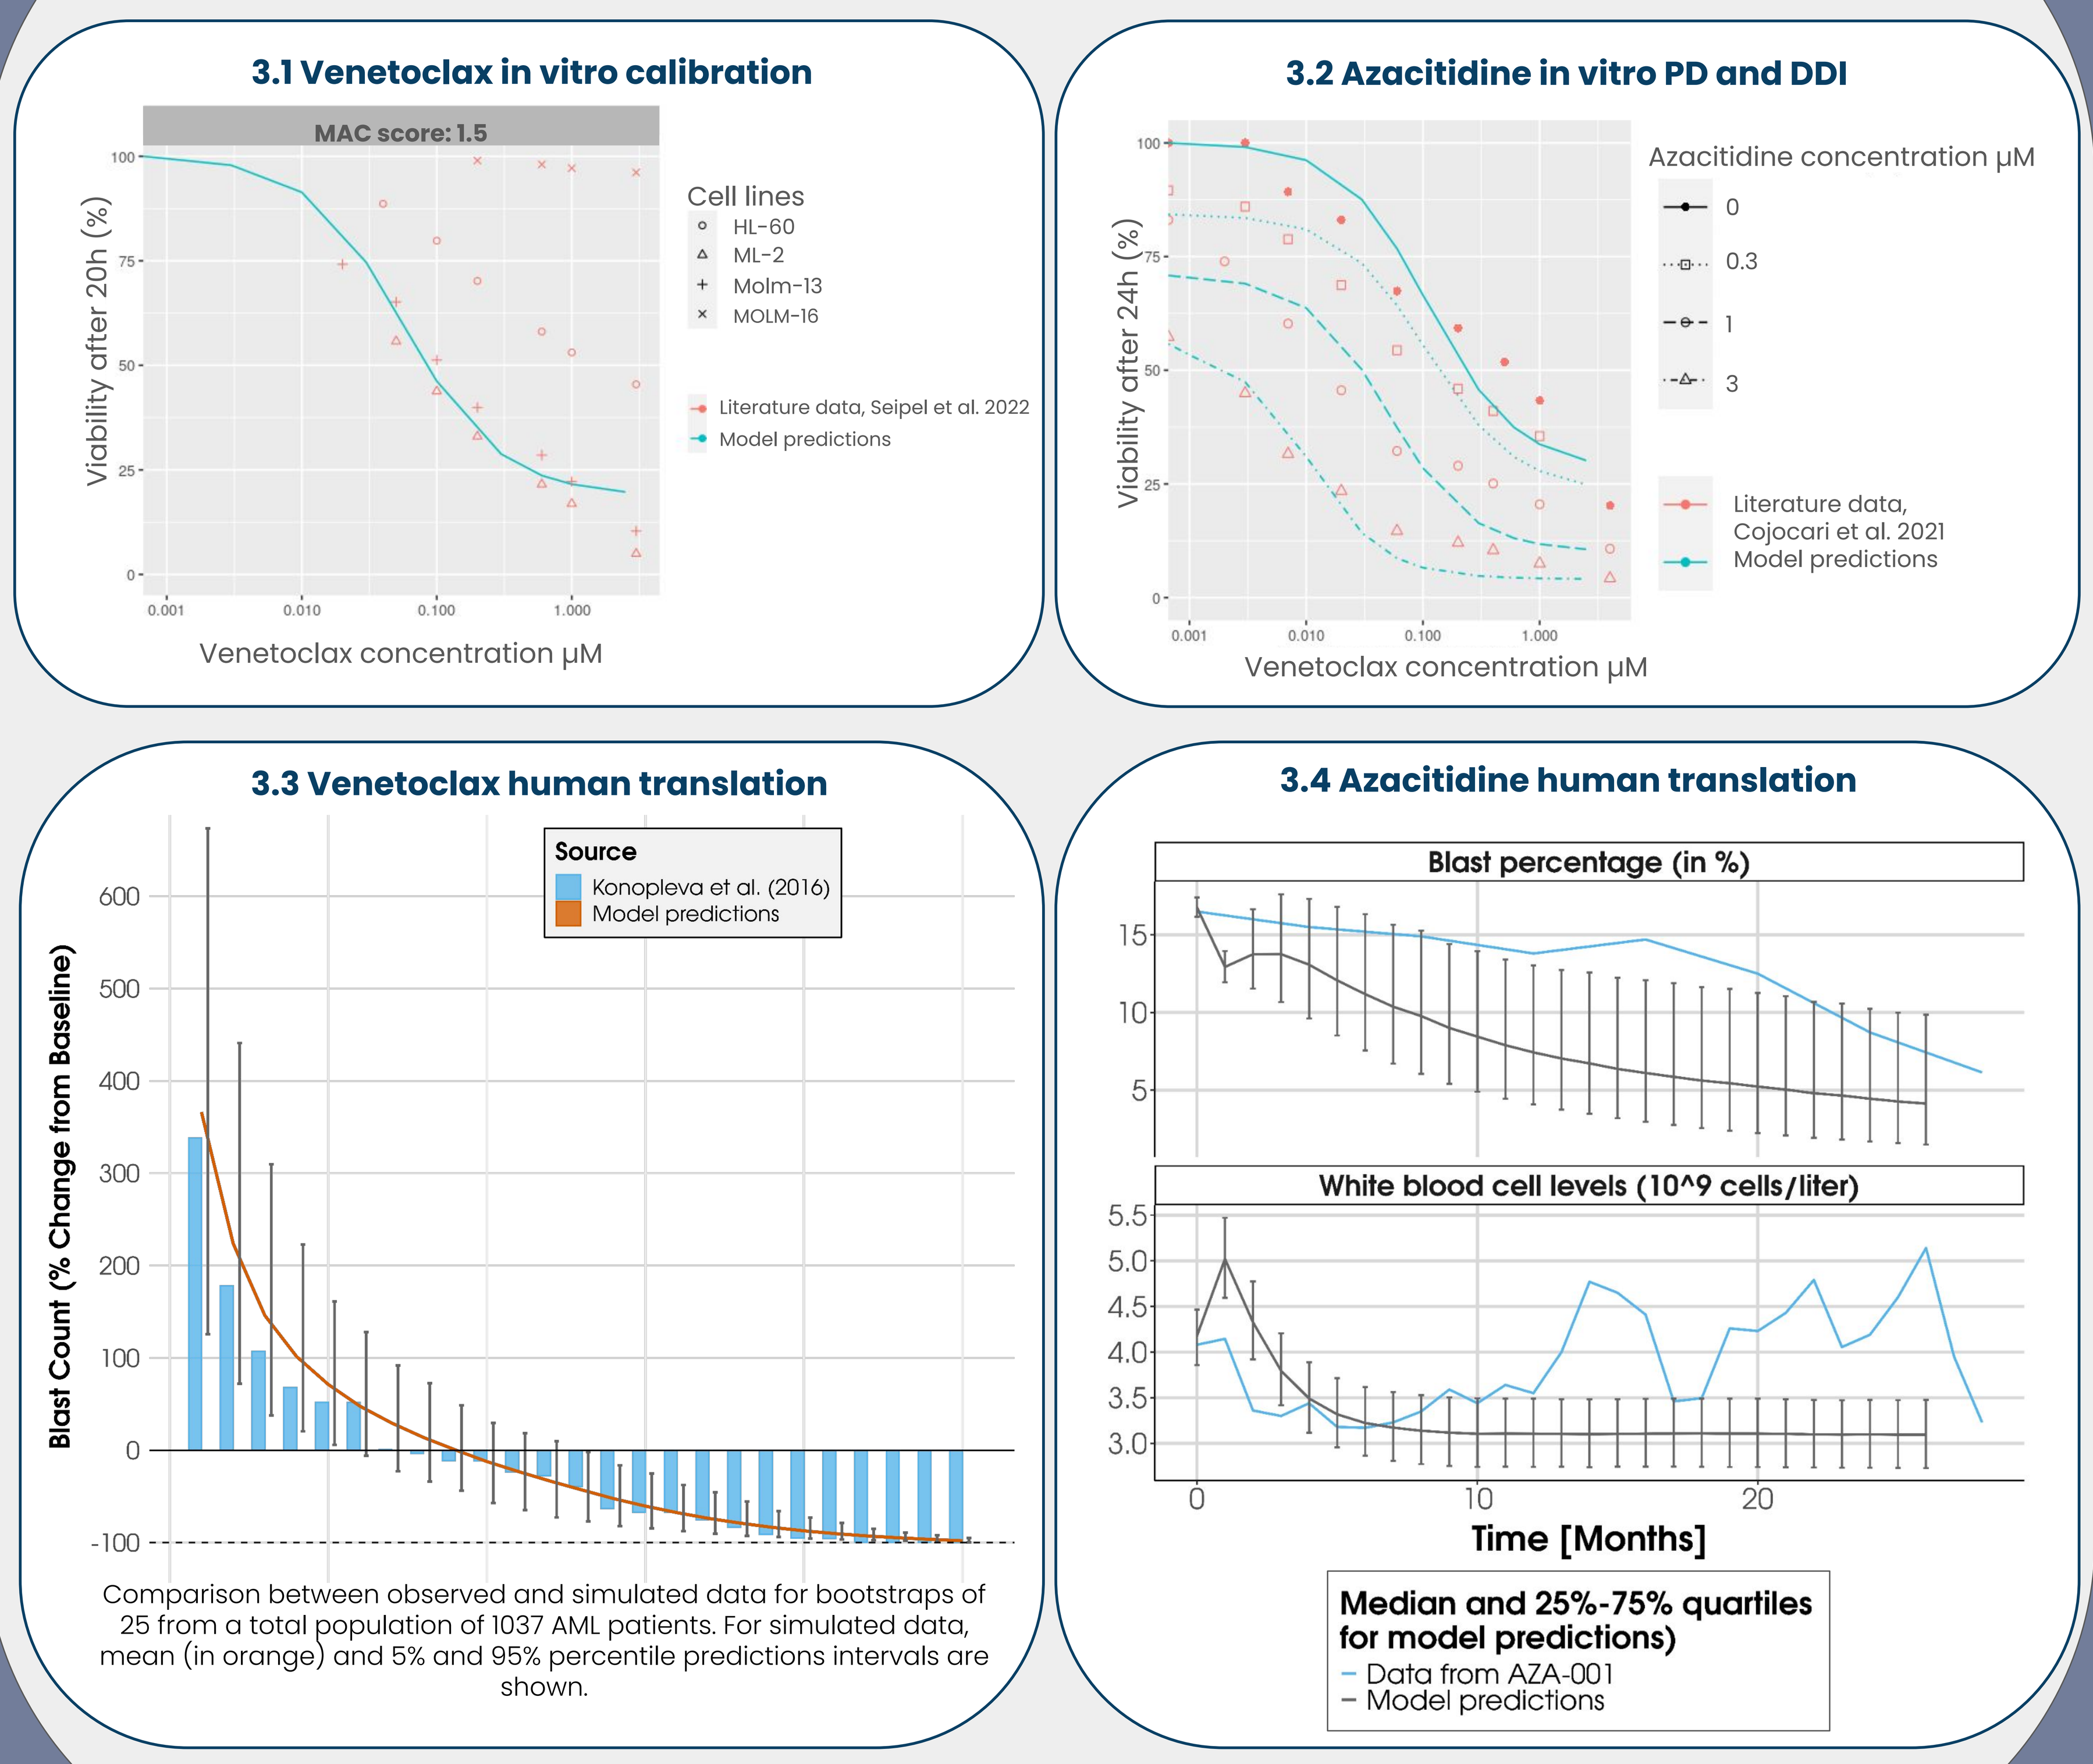
3.1 Venetoclax in vitro calibration
MAC score: 1.5
Viability after 20h (%)
Venetoclax concentration µM
Cell lines
HL-60
ML-2
Molm-13
MOLM-16
Literature data, Seipel et al. 2022
Model predictions
3.2 Azacitidine in vitro PD and DDI
Viability after 24h (%)
Venetoclax concentration µM
Azacitidine concentration µM
0
0.3
1
3
Literature data, Cojocari et al. 2021
Model predictions
3.3 Venetoclax human translation
Blast Count (% Change from Baseline)
Source
Konopleva et al. (2016)
Model predictions
Comparison between observed and simulated data for bootstraps of 25 from a total population of 1037 AML patients. For simulated data, mean (in orange) and 5% and 95% percentile predictions intervals are shown.
3.4 Azacitidine human translation
Blast percentage (in %)
White blood cell levels (10^9 cells/liter)
Time [Months]
Median and 25%-75% quartiles for model predictions)
Data from AZA-001
Model predictions

Figure 3: Calibration results for Venetoclax and Azacitidine in vitro (3.1, 3.2) and in clinical setting (3.3, 3.4)

METHODS

We developed a QSP model of VEN pharmacodynamics (PD), capturing cellular uptake, Bcl-2 binding, and intrinsic apoptosis, then combined it with a cell line model and an AZA PD model to simulate in vitro viability assays. By integrating a mechanistic model of AML pathophysiology and a pharmacokinetic (PK) model for both treatments, we created a comprehensive model for human AML treated with VEN-AZA. Sensitivity to Bcl-2 family members was derived from assumptions and clinical MAC score data [1], Firstly, an in vitro model was calibrated in Figures 3.1, 3.2.[2][3]. Secondly, PK curves for Vene and Aza were calibrated using clinical data and the remaining free PD parameters were informed with clinical blast change data [4][5](Figures 3.3, 3.4). Simulations were conducted using the jinkō QSP simulation platform.

RESULTS

Our model successfully captured the in vitro efficacy of VEN and AZA, both individually and in combination for different doses.

➔ Translation to human

More importantly, translating the model to a human AML context proved successful. We were able to simulate response variability observed in VEN monotherapy trials, such as changes in bone marrow blast count after four weeks of treatment. Similarly, the model effectively captured the long-term behavior seen in AZA monotherapy trials, including changes in mean blast and white blood cell counts.

➔ Response stratification

Furthermore, we conducted simulations combined with sensitivity analysis to explore model predictions in more detail. These simulations qualitatively agreed with clinical observations on how response markers depend on the MAC score, suggesting that the model can potentially be used to predict treatment outcomes based on a patient's Bcl-2 sensitivity profile.

Simulated maximum reduction of bone marrow blast count change from baseline for different MAC score subgroups

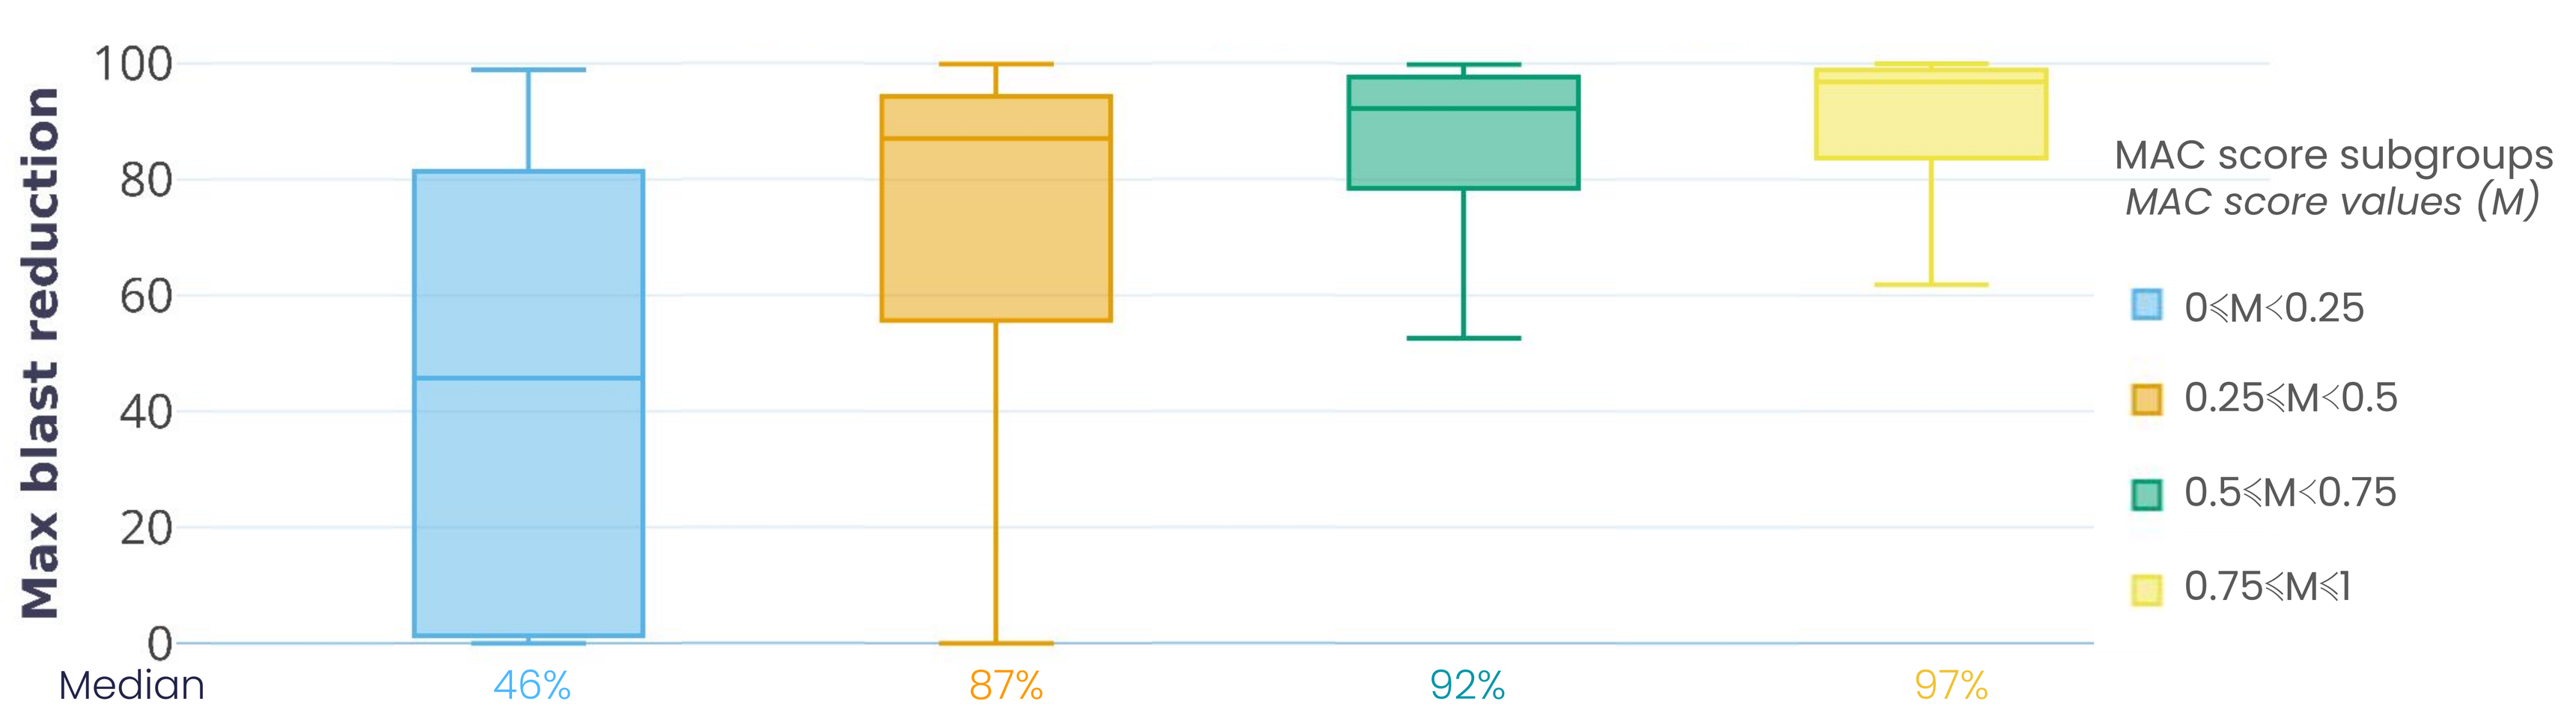
Max blast reduction
MAC score subgroups
MAC score values (M)
0⩽M<0.25
0.25⩽M<0.5
0.5⩽M<0.75
0.75⩽M⩽1
Median
46%
87%
92%
97%

Figure 4: Trial results comparing maximum reduction of bone marrow blast count change from baseline for different MAC score subgroups after Ven + Aza treatment. MAC Score represents the ratio of Bcl-2 expression over other anti-apoptotic protein expression (BCL-xL and MCL)

CONCLUSION

➔ This study presents a groundbreaking QSP model for VEN-AZA therapy. To our knowledge, this is the first model to:
- Reproduce in vitro and clinical trial data for both VEN and AZA as single agents.
- Derive response variability from Bcl-2 sensitivity data obtained in clinical settings.

➔ This work paves the way for developing a comprehensive AML QSP platform that can evaluate additional treatment combinations targeting the intrinsic apoptosis pathway. Such a platform could ultimately contribute to the development of personalized treatment strategies for AML patients, leading to improved clinical outcomes.

REFERENCES

[1] Waclawiczek et al. (2023), [2] Seipel et al. (2022), [3] Cojocari et al. (2021), [4] Konopleva et al. (2016), [5] PMDA of Japan (2017)

Detailed references available upon request.